PDB9

Long-term Clinical Outcomes of Exenatide Once-Weekly vs Insulin Glargine for the Treatment of Type 2 Diabetes Projected using the CORE Diabetes Model

Louise Timlin[1], Amélie Beaudet[2], Bernard Wilson[1], David Bruhn[3], Kristina S Boye[3], James L Palmer[2], Adam Lloyd[2]

[1]Eli Lilly and Company, Erl Wood, Surrey, UK; [2]IMS Health, Basel, Switzerland, [3]Eli Lilly and Company, Indianapolis, Indiana, USA

ABSTRACT

Objectives: This analysis projected the long-term incremental difference in clinical outcomes for exenatide once-weekly (EQW) compared with insulin glargine. A 26-week, randomised, clinical trial in 456 patients with type 2 diabetes failing treatment with oral antidiabetic agents (OAD) was performed to compare EQW to insulin glargine. EQW and insulin glargine were associated with (LS) mean decreases in HbA_{1c} (-1.47% and -1.31%), decreases in systolic blood pressure (-3.03mmHg and -0.63mmHg), and changes in body weight (-2.6kg and +1.4kg) from baseline, respectively.

Methods: The published and validated CORE Diabetes Model (CDM) was used to project clinical outcomes over patient lifetimes using the trial data. The model simulates disease progression by combining 15 Markov-based sub-models to determine the occurrence and time to onset of diabetes-related complications, life years gained and quality adjusted life years (QALYs). Standard CDM utility values for diabetes-related complications and body weight were included in the model. A discount rate of 3.5% was applied.

Results: Treatment with EQW compared to insulin glargine was associated with a higher projected life expectancy (11.93 vs 11.81 years) and quality-adjusted life expectancy (8.032 vs 7.849 *QALYs*). The projected cumulative incidence of all diabetes-related complications was lower for EQW compared to glargine except for stroke, amputation, cataract and peripheral vascular disease (PVD): Congestive Heart Failure (31.35% vs 32.54%), Stroke (29.66% vs 29.62%), Myocardial Infarction (20.61% vs 21.25%), Angina (15.77% vs 16.04%), PVD (12.80% vs 12.75%), Amputation Recurrent Ulcer (1.26% vs 1.18%). Moreover, EQW was associated with a longer projected mean time to onset of first complication versus glargine (4.80 versus 4.59 years).

Conclusions: Long term projections based on the findings of a recent randomised controlled trial indicate that EQW was more likely to improve life expectancy and quality-adjusted life expectancy, reduce complication rates and delay the time to onset of diabetes-related complications compared with insulin glargine.

Acknowledgement: This work was supported by an unrestricted grant from Eli Lilly and Company.

OBJECTIVES

- To project the long-term incremental difference in clinical outcomes for exenatide once-weekly (EQW) compared with insulin glargine.

METHODS

- The published and validated CORE Diabetes Model (CDM) was used to project clinical outcomes over patient lifetimes using trial data[1, 2].
- The model simulates disease progression by combining 15 inter-dependent Markov-based sub-models to determine the occurrence and time to onset of diabetes-related complications, life years gained and quality adjusted life years (QALYs).

Base case assumptions

Simulation cohort:

- Baseline characteristics were based on a 26 week, randomised, clinical trial in 456 patients with type 2 diabetes failing treatment with oral antidiabetics (OAD) alone, comparing the safety and efficacy of EQW 2.0mg plus OAD with that of insulin glargine plus OAD[3].
- EQW and insulin glargine were associated with (LS) mean decreases in HbA1c (-1.47% and -1.31%), decreases in systolic blood pressure (-3.03mmHg and -0.63mmHg), and changes in body weight (-2.6kg and +1.4kg) from baseline, respectively.
- Baseline diabetes complication rates were used from the NICE CG87 cohort[4,5]
- For diabetes complications not reported in the CG87 cohort, a baseline prevalence rate of zero was assumed.
- Table 1 presents the baseline demographics and risk factor status of patients included in the base case simulations.

Perspective of the analysis:

- The analysis has been conducted from the UK national health service payer perspective.

Time horizon and discounting:

- In accordance with NICE recommendations, a 50-year time horizon was used (mean baseline age of 57.92 years).
- Benefits were discounted at 3.5% annually in line with NICE recommendations.

Primary outcome:

- Quality adjusted life expectancy (QALE) was the primary outcome measure.

METHODS CONT.

Table 1. Patient characteristics

Characteristics	Value	Reference
HbA1c (%)	8.30	[3]
Age (years)	58	[3]
Male (%)	53.30	[3]
Ethnicity (proportion)		[3]
Caucasian	0.8310	
African-descent	0.0070	
Asian/Pacific-Islander	0.0590	
Hispanic	0.1030	
Duration of diabetes (years)	7.89	[3]
Systolic blood pressure (mmHg)	134.40	[3]
Baseline total cholesterol (mg/dL)	186.34	[3]
Baseline HDL-C (mg/dL)	46.25	[3]
Baseline LDL-C (mg/dL)	103.72	[3]
Baseline triglycerides (mg/dL)	162.61	[3]
Body mass index (kg/m^2)	32.27	[3]
Prevalent conditions	**Proportion**	**Reference**
Myocardial infarction	0.082	[5]
Peripheral vascular disease	0.0	Assumed
Stroke	0.049	[5]
Congestive heart failure	0.037	[5]
Angina	0.0	Assumed
Background diabetic retinopathy	0.177	[5]
Proliferative diabetic retinopathy	0.0	Assumed
Macular oedema	0.0	Assumed
Cataract	0.0	Assumed
Amputation	0.0	Assumed
Neuropathy	0.065	[5]

HDL-C: high density lipoprotein cholesterol; LDL-C: low density lipoprotein cholesterol

Treatment pathway:

- Due to the progressive nature of Type 2 Diabetes, patients treated with EQW were switched to insulin glargine after five years of treatment.

Treatment associated utilities:

- Utilities associated with weight change and nausea were included in the annual utility scores associated with treatment[6,7]

Treatment effects:

- Treatment effects, presented in table 2, were drawn from a 26 week, randomised, clinical trial[3].
- The intervention effects are applied in the first year of treatment.

Table 2. Treatment effects applied in CDM

	Mean change from baseline ± SD	
	Exenatide Once-Weekly	**Insulin Glargine**
HbA1c (%)	-1.47±0.76	-1.31±0.90
Systolic blood pressure (mmHg)	-3.03±16.57	-0.63±14.88
Total cholesterol (mg/dL)	-4.64±35.41	-1.55±34.64
HDL-cholesterol (mg/dL)	0.00±5.90	0.39±5.77
LDL-cholesterol (mg/dL)	-1.93±29.51	1.55±28.87
Triglycerides (mg/dL)	-11.52±101.89	-15.95±112.52
BMI (kg/m^2)	-0.93±1.08	0.51±1.07
Major hypoglycaemia (events/100 patient years)	0	0
Minor hypoglycaemia (events/100 patient years)	43	127

BMI: Body Mass Index; HbA1c: glycosylated haemoglobin; HDL: high density lipoprotein; LDL: low density lipoprotein; SD: standard deviation; SBP: systolic blood pressure

RESULTS

- Treatment with EQW was projected to increase life-expectancy and quality adjusted life expectancy versus treatment with insulin glargine (Table 3)
- As shown in table 3, projected life expectancy increased by 0.117 years with EQW (11.925 and 11.808 years for EQW and insulin glargine) and was associated with projected quality adjusted life expectancy of 8.032 versus 7.849 QALYs for EQW and insulin glargine.

Table 3. Basecase results

	Exenatide QW	Insuline Glargine	Difference
Quality-adjusted life expectancy (QALYs)	8.032 (0.108)	7.849 (0.112)	0.183 (0.150)
Life expectancy (years)	11.925 (0.156)	11.808 (0.162)	0.117 (0.217)
Undiscounted life expectancy (years)	17.076 (0.271)	16.856 (0.285)	0.220

Exenatide QW: exenatide once weekly; QALY: quality-adjusted life years; Values shown are means with standard deviations in parentheses.

RESULTS CONT.

- Treatment with EQW was associated with a longer projected mean time to onset of first complication versus insulin glargine (4.80 versus 4.59 years) (Table 4)
- The projected onset of most diabetes related complications was delayed by two or three months with EQW versus insulin glargine (Table 4).

Table 4. Time alive and free of complication

	Time alive and free of complication (years)		
Complication	**Exenatide QW**	**Insulin Glargine**	**Difference**
Any complications	4.80	4.59	0.21
Background retinopathy	12.30	12.07	0.23
Proliferative retinopathy	16.57	16.35	0.22
Microalbuminuria	14.08	13.79	0.29
Gross proteinuria	16.72	16.48	0.24
End-stage renal disease	17.06	16.84	0.22
First ulcer	15.76	15.55	0.21
Amputation	16.80	16.58	0.22
Neuropathy	11.07	10.75	0.32
Peripheral vascular disease	16.07	15.89	0.18
Congestive heart failure	15.27	14.98	0.29
Angina	15.73	15.51	0.22
Myocardial infarction	15.37	15.15	0.22
Stroke	14.84	14.63	0.21
Cataract	16.00	15.79	0.21
Macular oedema	15.45	15.16	0.29
Severe vision loss	16.41	16.16	0.25

Exenatide QW: exenatide once weekly; Values are shown in means.

- The projected cumulative incidence of all diabetes-related complications (see Figure 1) was lower for EQW compared to glargine (except for stroke, amputation, cataract and peripheral vascular disease (PVD) due to the longer life expectancy for the EQW treatment arm): Congestive Heart Failure (31.35% vs 32.54%), Stroke (29.66% vs 29.62%), Myocardial Infarction (20.61% vs 21.25%), Angina (15.77% vs 16.04%), PVD (12.80% vs 12.75%), Amputation Recurrent Ulcer (1.26% vs 1.18%).

Figure 1. Cumulative incidence of complications

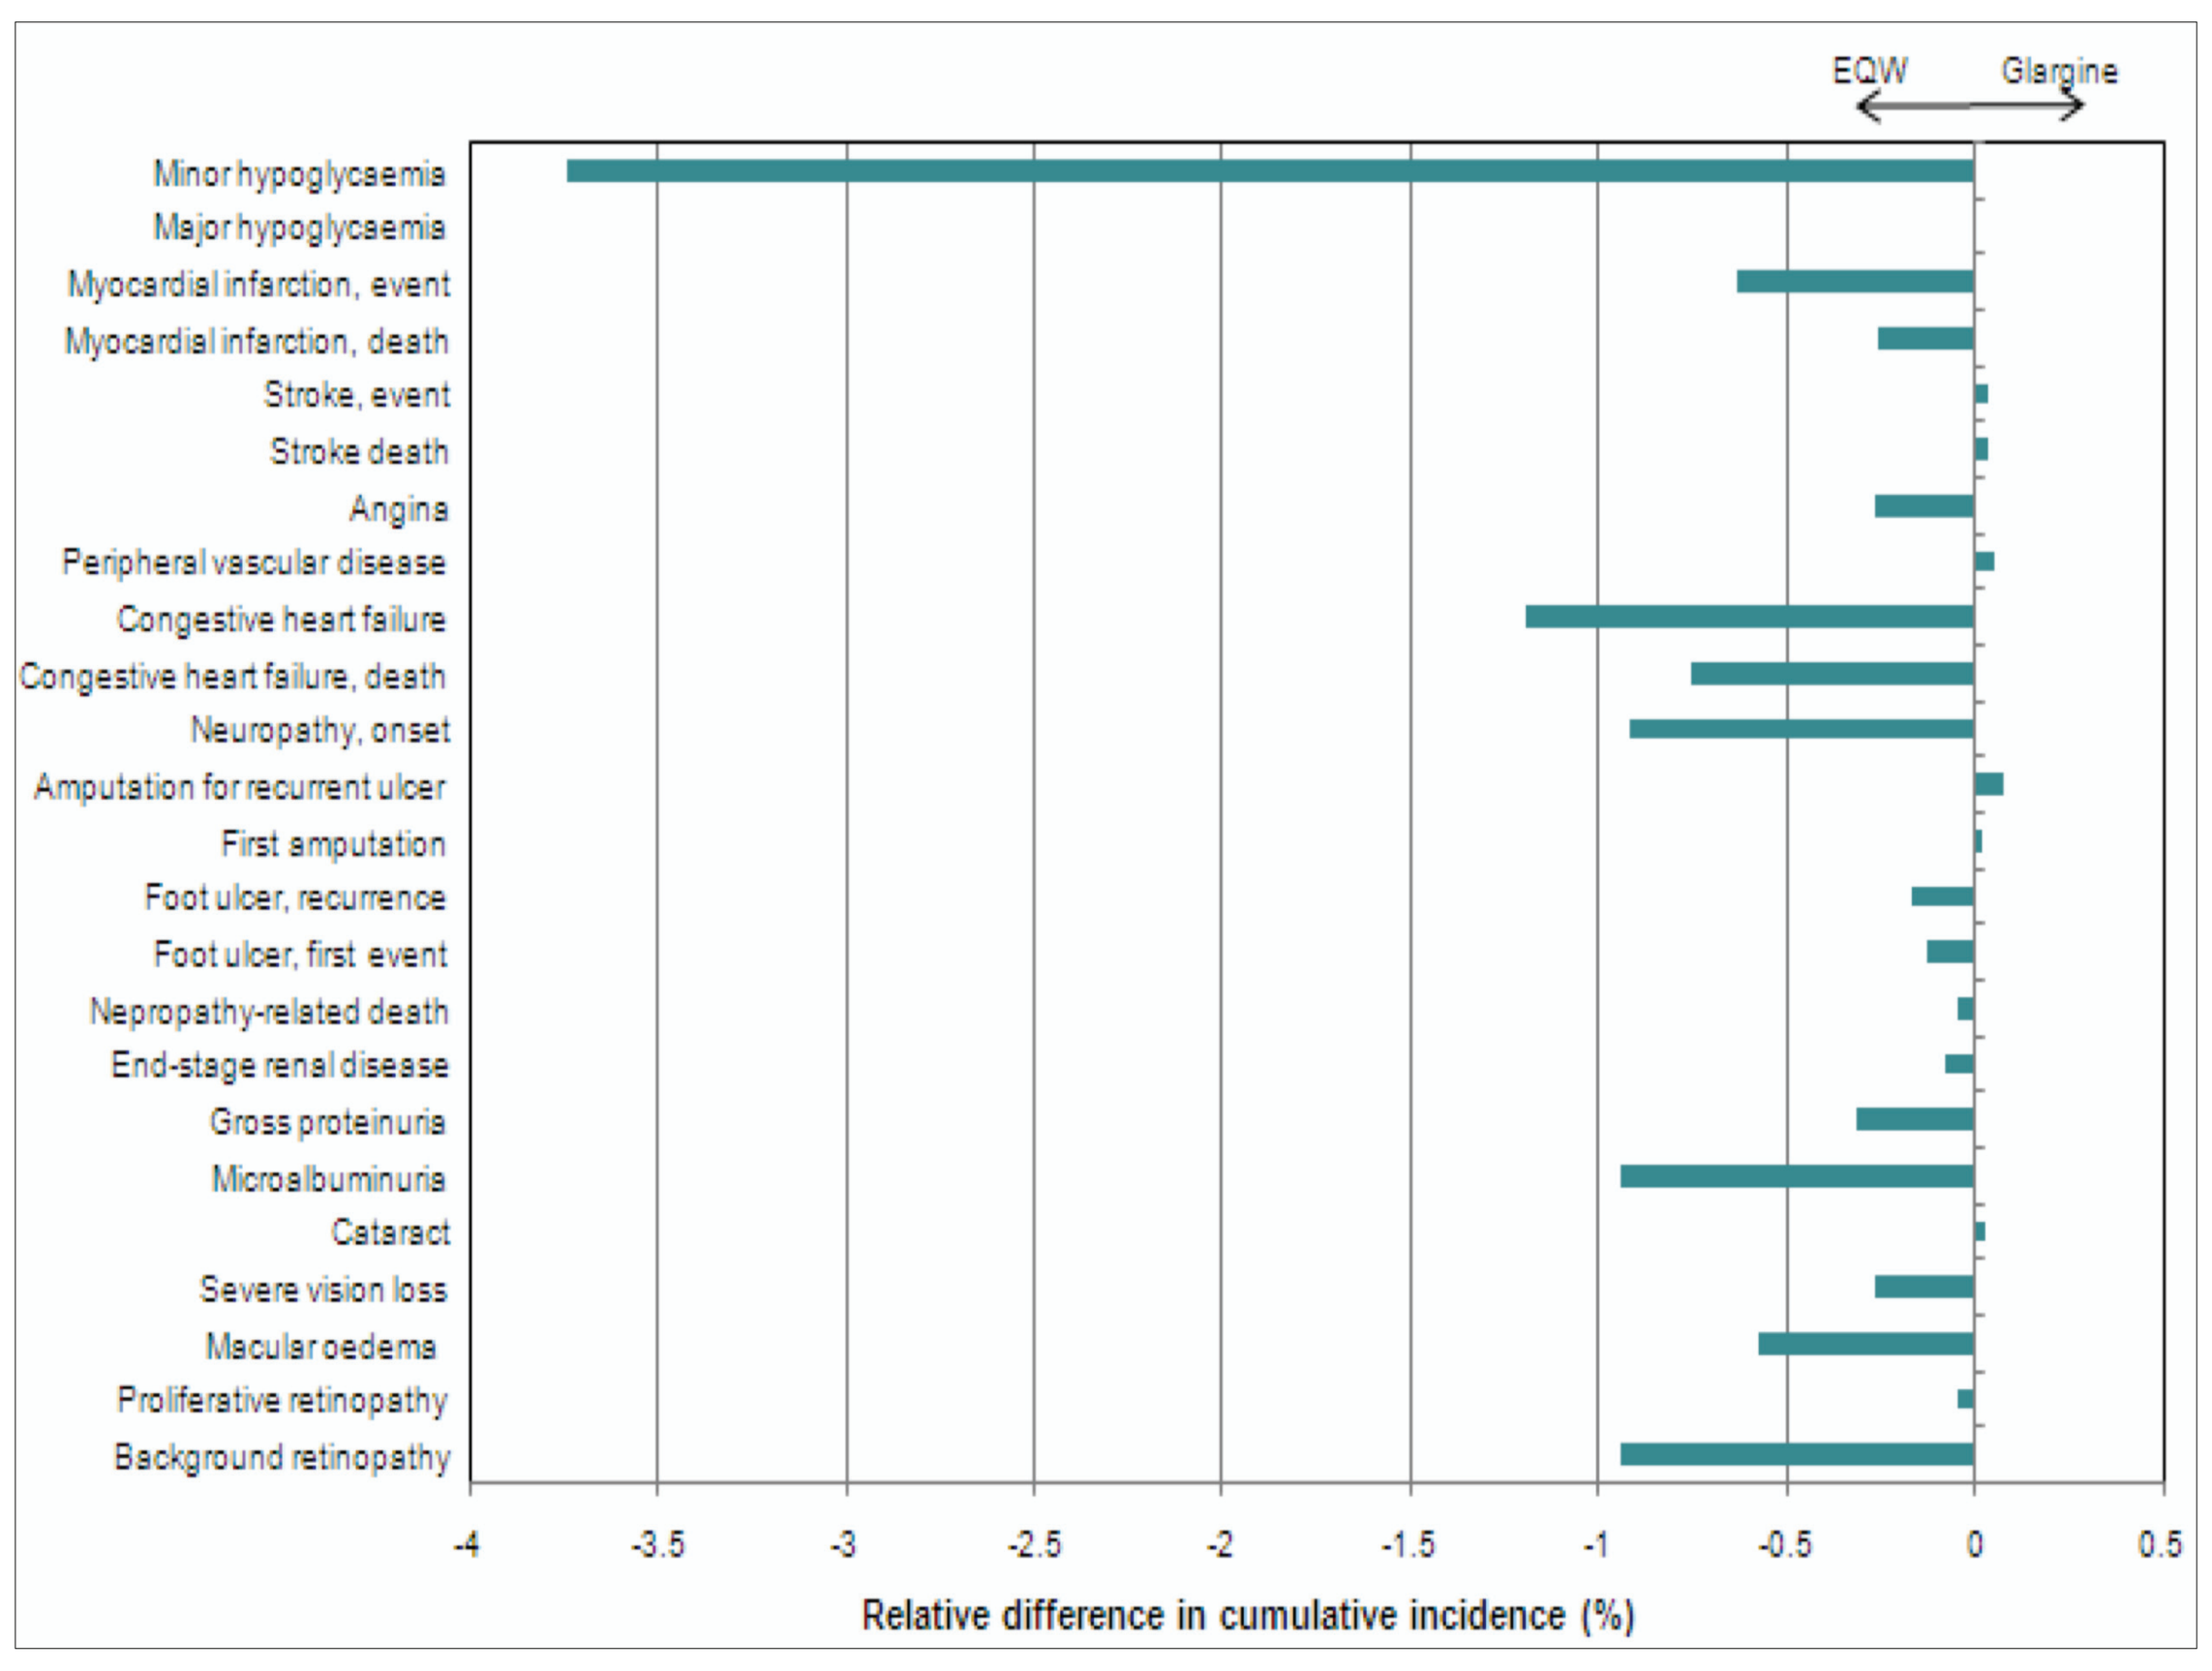
EQW
Glargine
Minor hypoglycaemia
Major hypoglycaemia
Myocardial infarction, event
Myocardial infarction, death
Stroke, event
Stroke death
Angina
Peripheral vascular disease
Congestive heart failure
Congestive heart failure, death
Neuropathy, onset
Amputation for recurrent ulcer
First amputation
Foot ulcer, recurrence
Foot ulcer, first event
Nepropathy-related death
End-stage renal disease
Gross proteinuria
Microalbuminuria
Cataract
Severe vision loss
Macular oedema
Proliferative retinopathy
Background retinopathy
-4
-3.5
-3
-2.5
-2
-1.5
-1
-0.5
0
0.5
Relative difference in cumulative incidence (%)

LIMITATIONS

- Patients were switched to insulin glargine after five years.
- Intermediate end points (HbA1c, BP, lipids, BMI) were used to project long term outcomes.
- Intervention effects were applied in the first year only.

CONCLUSIONS

- In the UK setting, exenatide QW was associated with a projected greater improvement in long-term clinical outcomes compared to insulin glargine.
- In long-term projections over a 50-year time horizon, based on the findings of a recent randomised controlled trial, EQW compared to insulin glargine was more likely to:
 - improve life expectancy and quality-adjusted life expectancy
 - reduce the cumulative incidence of complications (except for stroke, amputation, cataract and PVD)
 - delay the time to onset of diabetes-related complications.

Acknowledgments: This work was supported by an unrestricted grant from Eli Lilly and Company.

References:

[1]Palmer AJ et al. The CORE Diabetes Model: projecting long-term clinical outcomes, costs and cost-effectiveness of interventions in diabetes mellitus (types 1 and 2) to support clinical and reimbursement decision-making. Curr Med Res Opin 2004a Aug;20(Suppl 1):5-26.
[2]Palmer AJ, et al. Validation of the CORE Diabetes Model against epidemiological and clinical studies. Curr Med Res Opin 2004b;20(Suppl 1):S27-S40.
[3]Eli Lilly and company. GWBR Clinical Study Report. 2009.
[4]National Institute for Health and Clinical Excellence. Guide to the methods of technology appraisal. National Institute for Health and Clinical Excellence 2008 January 6 Available from: URL: http://www.nice.org.uk/ media/B52/A7/TAMethods GuideUpdatedJune2008.pdf
[5]National Institute for Health and Clinical Excellence. Clinical guidelines CG87. 2009 Jan 5.
[6]Bagust A, Beale S. Modelling EuroQol health-related utility values for diabetic complications from CODE-2 data. Health Econ 2005 Mar;14(3):217-30.
[7]Matza LS, et al. Utilities and disutilities for type 2 diabetes treatment-related attributes. Qual Life Res 2007 Sep;16(7):1251-65.

ISPOR 13th Annual European Congress; Prague, Czech Republic; November 6-9, 2010

Sponsored by Eli Lilly and Company *{Exen-00038957}*

eISPOR DUR 3 CORE model poster.indd 1

29/10/10 06:40:50